Pooled data analysis of the safety and tolerability of intravenous pelareorep in combination with chemotherapy in 500+ cancer patients

A. A. Gutierrez, C. Reid, A. Dzugalo, M. Crawford, K. Cheetham, M. Parsi, A. Penman, N. Noronha, D. Galindez, R. O'Flynn, M. Coffey

Oncolytics Biotech Inc
Calgary, Canada

BACKGROUND

Pelareorep (REOLYSIN®,**R**) an unmodified **reovirus** Type 3 Dearing (T3D) strain, is a systemically delivered immuno-oncolytic virus (IOV) being investigated in solid tumors and hematological malignancies. **R**'s anti-tumor activity is based on two modes of action (Figure 1):

1) Selective viral replication in permissive cancer cells which leads to tumor cell lysis
2) Anti-Tumor Immunity
 - Activation of innate immunity in response to infection results in a cascade of chemokines/cytokines causing NK cells to be activated and attack cancer cells[1]
 - A specific adaptive immune response triggered by tumor-and viral-associated antigens displayed by antigen- presenting cells (APCs), infected tumor cells and/or dendritic cells, to T-cells[2]

Figure 1. Pelareorep's Mechanism of Action

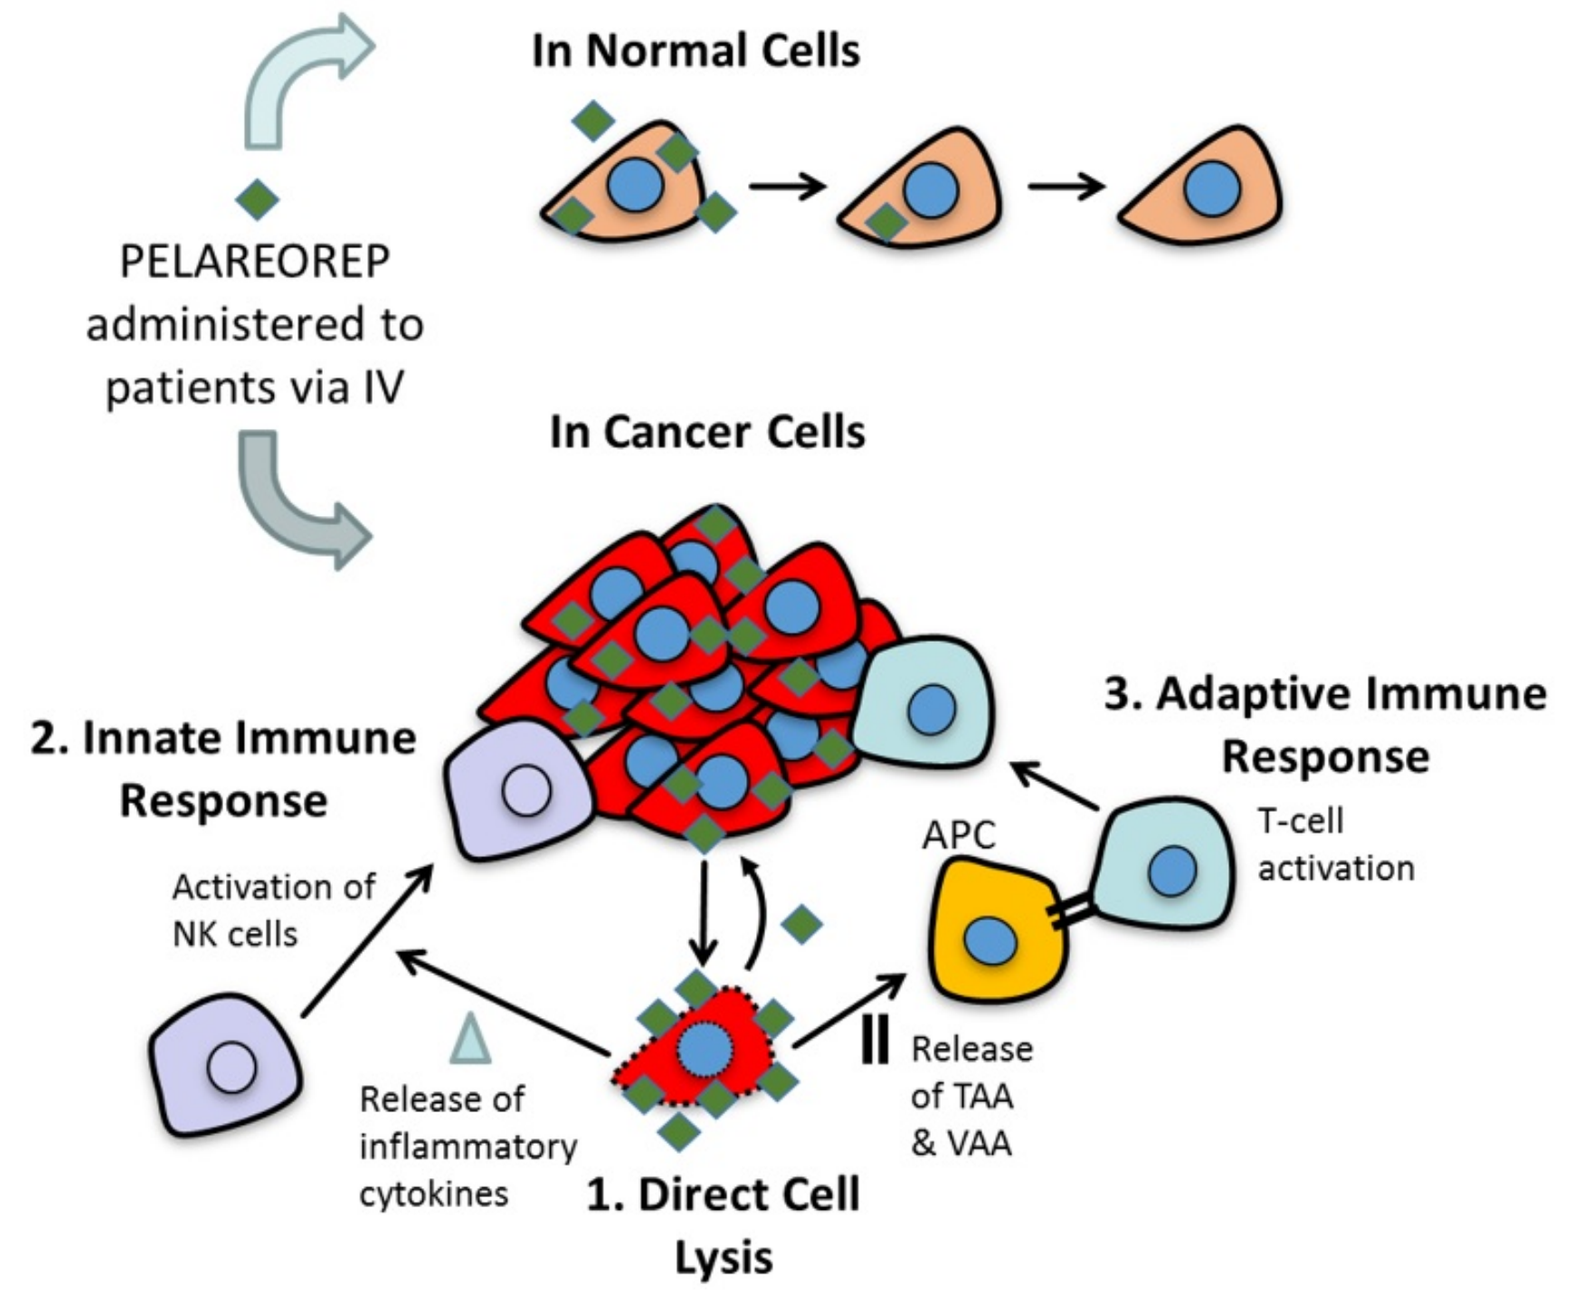
In Normal Cells
PELAREOREP administered to patients via IV
In Cancer Cells
2. Innate Immune Response
Activation of NK cells
3. Adaptive Immune Response
T-cell activation
APC
Release of inflammatory cytokines
1. Direct Cell Lysis
Release of TAA & VAA

1420 patients have been enrolled in clinical studies in the US, Canada and EU. Of these, 1029 patients have received **R** intravenously (n=938) or by intratumoral injections (n=91). The principal adverse effects in monotherapy studies with IV **R** in solid tumors are encompassed in the term "flu-like syndrome" and include Grade 1 and 2 signs and symptoms such as fever, chills, headache, fatigue, rhinorrhea, cough, sweating, myalgia, nausea, vomiting, and/or diarrhea[3]. A recent phase 2 study conducted by the Canadian Clinical Trials Group (CCTG) showed that the addition of paclitaxel (**P**) to **R**, Arm A, improved overall survival (17.4m) vs. **P** alone, Arm B, (10.4m) in metastatic breast cancer patients (HR 0.65, 80% CI 0.46-0.91, p=0.1)[4] (Figure 2A). A post-hoc analysis conducted by Cytel (on behalf of Oncolytics), suggests that the survival benefit with **R** + **P** may be more significant in hormone receptor-positive patients (ER+/PR+). In this subgroup (n=47) the overall survival with **R** was 21.8m vs. 10.8 m with **P** alone (HR 0.36, p=0.003) (Figure 2B). A pooled analysis was thus conducted to better characterize **R**'s safety profile in combination regimens with **P**.

METHODS

Nine trials with paclitaxel (**P**), paclitaxel + pelareorep (**PR**), carboplatin + paclitaxel (**CP**) or carboplatin + paclitaxel + pelareorep (**CPR**) were pooled. Standard doses of **P** (80mg/m² weekly on days 1,8,15 q28 days) or **CP** (carboplatin AUC 5-6 mg/mL·min plus paclitaxel 175-200 mg/m² q21 days) were administered. **R** was administered IV at a dose of $3x10^{10}$ $TCID_{50}$, 5-6 times in 21-28-day cycles.

Safety data for **P** ± **R** were pooled from 2 studies: advanced or metastatic breast[4] and ovarian cancer. Safety data for **CP** ± **R** were aggregated from 7 studies: advanced malignancies[5], head and neck cancer[6](2 studies), NSCL cancer[7], melanoma, SCCL[8], and pancreatic cancer[9].

To generate comparable tables across multiple studies as well as pooled summaries for selected variables, parameters were assessed for compatibility and mapped to a common variable for analysis. Demographic variables requiring adjustment to create homogeneous datasets were age, race, performance status and prior anti-cancer therapies (e.g. only integer portions of age were retained; performance status scales other than ECOG [e.g. Karnofsky] were mapped to ECOG categories; prior anti-cancer therapies such as bone marrow transplant, drug and/or immunotherapy, hematopoietic stem cell transplantation, and vaccine were mapped to immuno/hormonal/biological/targeted therapy category).

AEs were summarized by preferred term (PT), relationship to treatment, seriousness, and severity. Seriousness was not reported for AEs in NCI studies only. All other parameters were provided for each AE in all studies. Though PTs were assigned according to various versions of MedDRA (REOLYSIN® studies) or other similar coding dictionaries (NCI and CCTG studies), no adjustments were made other than for UK vs US spelling. Relationship to study treatment was categorized by mapping all relatedness categories to either related or not related. For studies where relationship was categorized by treatment agent, an AE related to any component of study treatment (**C**, **P**, or **R**) was counted as related to study treatment. No changes to severities were required. AEs were considered treatment emergent if they started on or after first study treatment up to 30 days post last study treatment. If incomplete date data rendered an AE ambiguous with respect to treatment emergence, the AE was included.

Figure 2. IND.213 Breast Cancer Study Overall Survival. (A) ITT Population, n=74 and (B) ER+/PR+, n=47

(A)

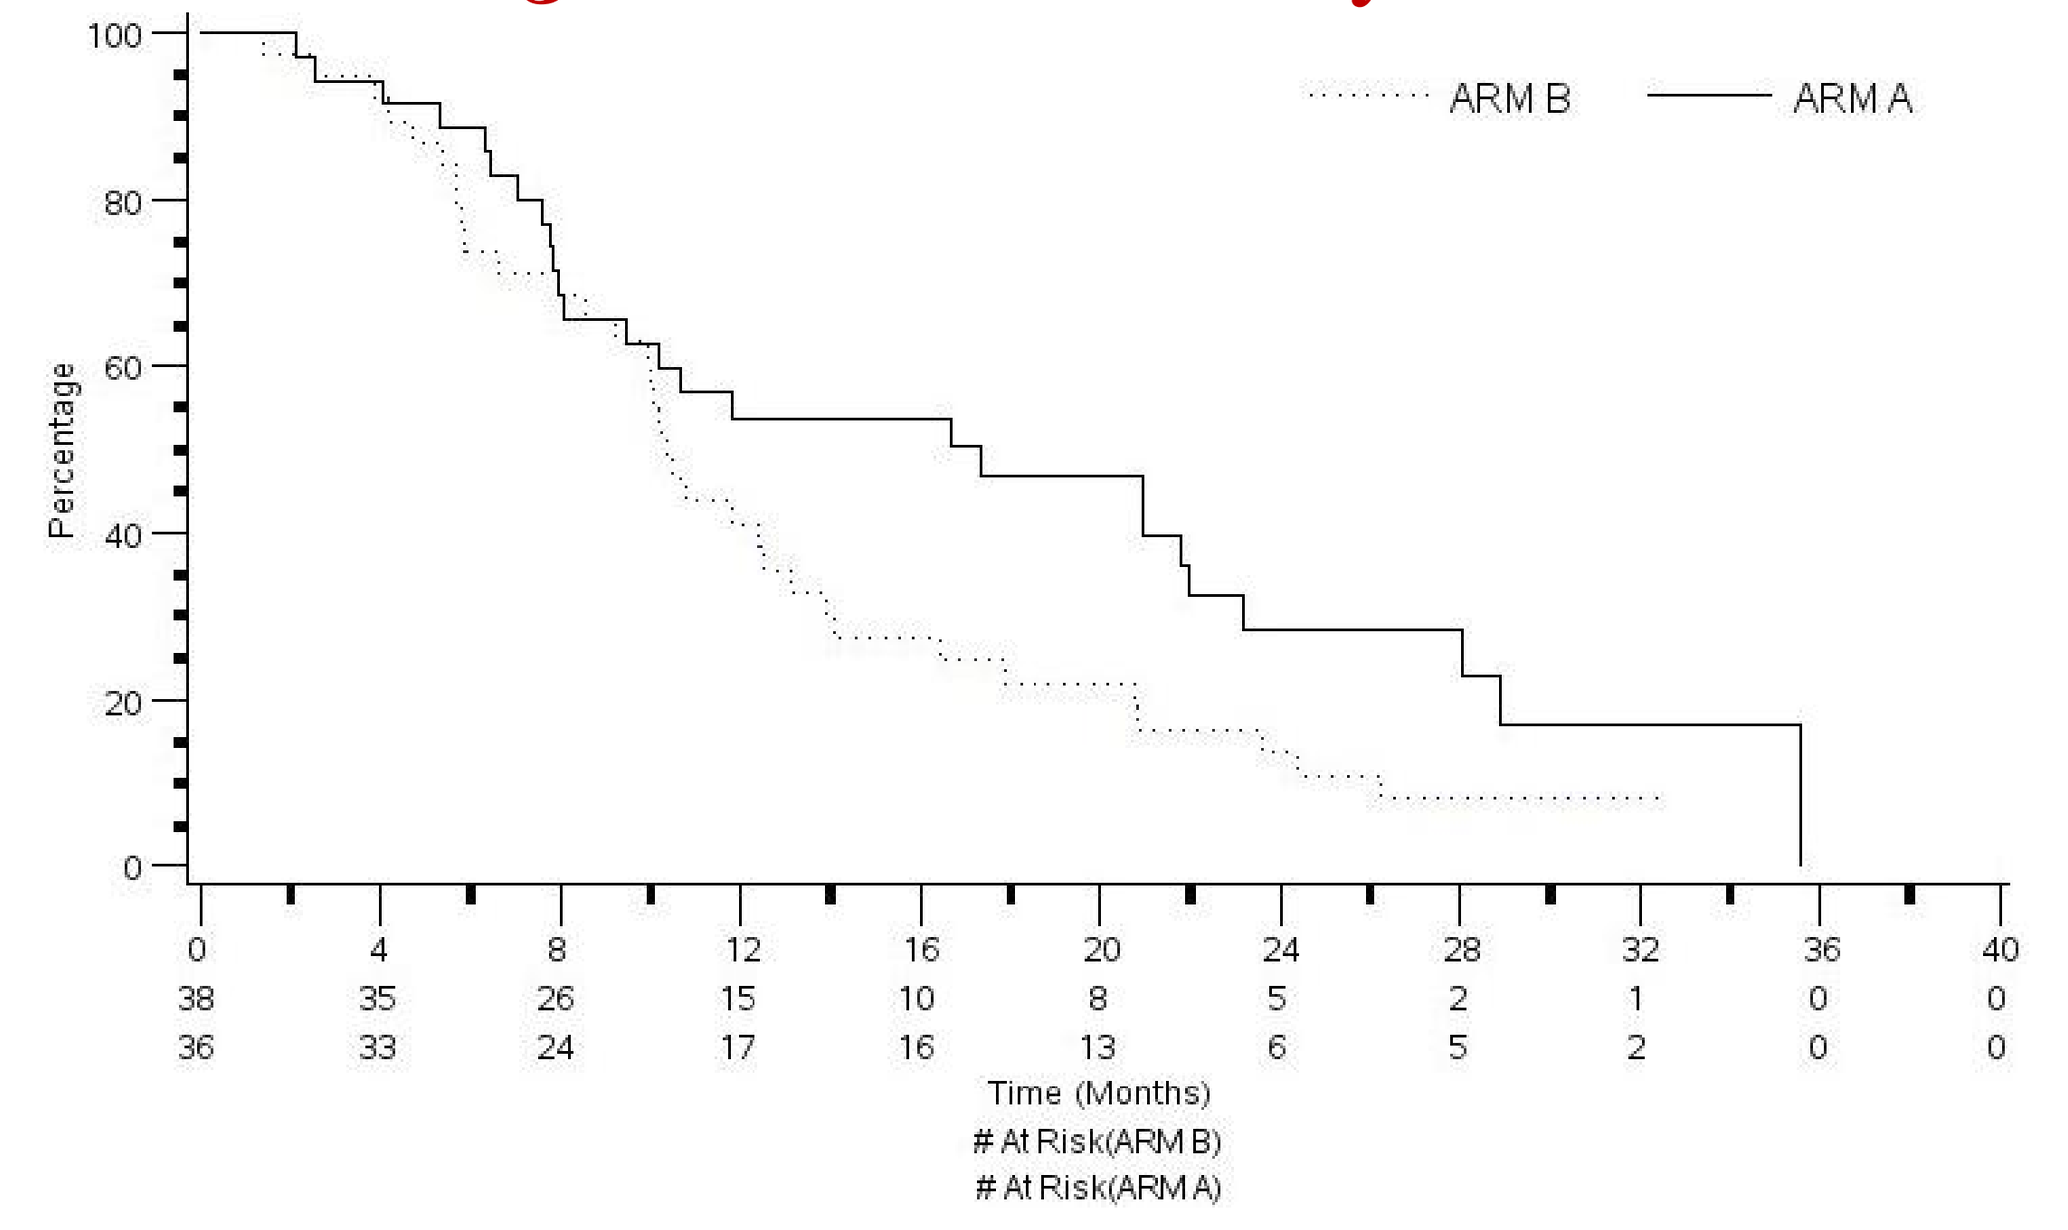
ARM B
ARM A
Percentage
100
80
60
40
20
0
0 4 8 12 16 20 24 28 32 36 40
38 35 26 15 10 8 5 2 1 0 0
36 33 24 17 16 13 6 5 2 0 0
Time (Months)
At Risk(ARM B)
At Risk(ARM A)

(B)

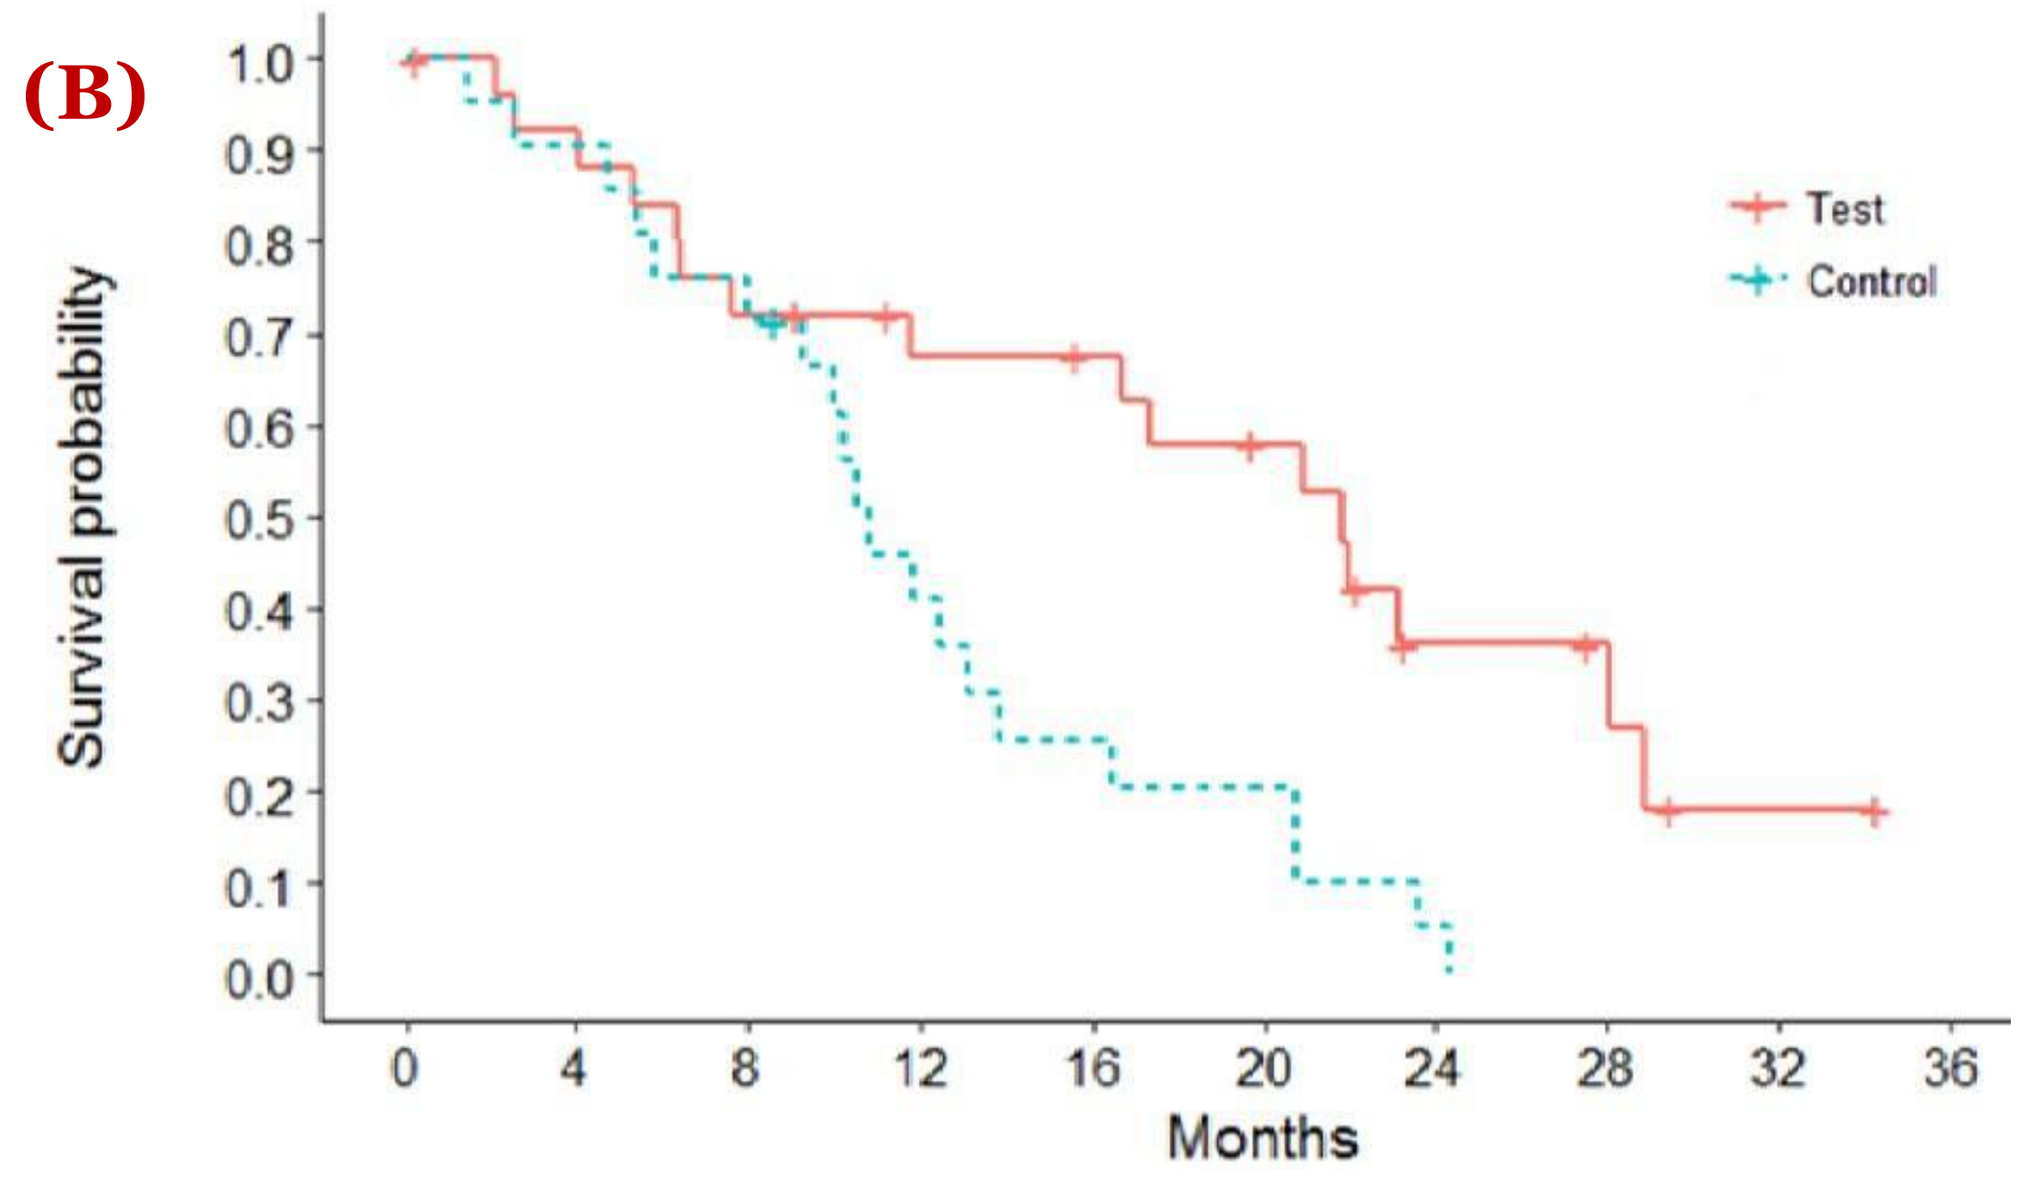
Test
Control
Survival probability
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0 4 8 12 16 20 24 28 32 36
Months

RESULTS

Demographics

547 patients; median age (59-62 y) and ECOG 0-1 status (90-97%) similar across the groups (Table 1). Studies with **P±R** included female patients only (except for 1 male), whereas male patients were the majority in the studies with **CPR** or **CP** (68-72%). All patients in **P** or **PR** had received prior chemo but only 26% in **CP** and 40% in **CPR**.

Table 1. Demographics Data (Safety Population)

	Studies with Paclitaxel+/- Pelareorep IV		Studies with Carboplatin + Paclitaxel +/- Pelareorep IV	
	PR (n= 95)	P (n= 86)	CPR (n= 248)	CP (n= 118)
Age (years)				
Median	62	59	60	61
Range	40, 87	35, 81	23, 84	23, 81
Gender: n (%)				
Female	95 (100)	85 (98.8)	80 (32.3)	33 (28.0)
Male	0	1 (1.2)	168 (67.7)	85 (72.0)
ECOG: n (%)				
0	51 (53.7)	42 (48.8)	93 (38.0)	37 (31.4)
1	41 (43.2)	28 (44.2)	130 (53.1)	69 (58.5)
2	3 (3.2)	6 (7.0)	21 (8.6)	12 (10.2)
Prior Therapy: n (%)				
Surgery	90 (94.7)	83 (96.5)	173 (69.8)	58 (49.2)
Radiation	36 (37.9)	35 (40.7)	139 (56.0)	70 (59.3)
Immuno/hormonal/ biological/targeted	28 (29.5)	35 (40.7)	17 (6.9)	0
Chemotherapy	95 (100)	86 (100)	100 (40.3)	31 (26.3)
Other	11 (11.6)	6 (7.0)	NA	NA

Note: Safety Population includes all patients treated with any study medication.
Note: Percentages are based on the number of subjects with non-missing data.
NA: Not applicable because data not collected in this study.

Exposure

Up to 55 cycles of **R** were administered (range 8 – 55 cycles), where one cycle consisted of **R** administered on Days 1-5, q21 days in all REO studies and NCI 8601; **R** administered on Days 1-5 q28 days in NCI GOG 0186H; and **R** administered on Days 1, 2, 8, 9, 15, 16 q28 days in CCTG i213). Duration of **R** treatment across all 9 studies ranged from 1 to 1216 days. Approximately 70% of **PR** patients and 88% of **CPR** patients received up to 6 months of **R** treatment.

Studies with Paclitaxel ± Pelareorep

Select Grade 1 and 2 AEs were more frequent in **R**-containing arms than in the control arms (**PR** vs. **P**, range in %), including: fever (65-46 vs. 11-8), chills (62-40 vs. 11-4), fatigue (64-40 vs. 67-21), flu-like symptoms (35-30 vs. 11-4), dyspnea (40-35 vs. 25-16) and the GI-related AEs of nausea (73-35 vs. 50-45), diarrhea (44-40 vs. 23-11) and vomiting (37-35 vs. 23-18).

Grade 3 and 4 laboratory abnormalities of white blood cell decreased and neutrophil count decreased were more common in the **R**-containing arms. Fever, hypotension and what was coded as "febrile neutropenia" may also reflect the effect of **R**, but the incidence of sepsis was similar for both groups (Table 2).

Table 2. Pooled Grade 3 and 4 Treatment Emergent Adverse Events Reported for ≥ 3% of Pelareorep-Treated Patients (2 studies): Paclitaxel ± Pelareorep

	Pelareorep IV Dose Group with Paclitaxel 80 mg/m² ($TCID_{50}$)			
Preferred Term	CCTG I213 Pelareorep (N=43) n (%)	NCI GOG-0186 Pelareorep (N=52) n (%)	CCTG I213 No Pelareorep (N=38) n (%)	NCI GOG-0186 No Pelareorep (N=48) n (%)
White blood cell decreased	0	17 (32.7)	0	3 (6.3)
Neutrophil count decreased	0	15 (28.8)	0	5 (10.4)
Hypertension	0	12 (23.1)	1 (2.6)	6 (12.5)
Anemia	0	10 (19.2)	0	7 (14.6)
Fatigue	2 (4.7)	8 (15.4)	3 (7.9)	5 (10.4)
Dyspnea	1 (2.3)	6 (11.5)	0	1 (2.1)
Lung infection	2 (4.7)	4 (7.7)	0	1 (2.1)
Hypoalbuminemia	0	5 (9.6)	0	4 (8.3)
Hyponatremia	0	5 (9.6)	0	2 (4.2)
Abdominal Pain	0	4 (7.7)	0	6 (12.5)
Ascites	0	4 (7.7)	0	4 (8.3)
Hypotension	1 (2.3)	3 (5.8)	0	2 (4.2)
Nausea	1 (2.3)	3 (5.8)	2 (5.3)	4 (8.3)
Small intestinal obstruction	0	4 (7.7)	0	6 (12.5)
Syncope	1 (2.3)	3 (5.8)	0	0
Enterocolitis infectious	1 (2.3)	2 (3.8)	0	0
Fever	1 (2.3)	2 (3.8)	1 (2.6)	0
Hypocalcemia	0	3 (5.8)	0	2 (4.2)
Hypoxia	0	3 (5.8)	0	0
Pleural Effusion	0	3 (5.8)	0	0
Sepsis	0	2 (3.8)	1 (2.6)	2 (4.2)
Atrial fibrillation	0	2 (3.8)	0	0
Diarrhea	0	2 (3.8)	3 (7.9)	4 (8.3)
Vomiting	0	1 (1.9)	3 (7.9)	4 (8.3)
Thromboembolic Event	0	2 (3.8)	1 (2.6)	3 (6.3)
Dehydration	1 (2.3)	0	1 (2.6)	4 (8.3)
Lymphocyte count decreased	0	1 (1.9)	0	3 (6.3)

Serious TEAEs considered to be treatment-related by the Investigator are presented in Table 3. Fever, nausea, and vomiting were the only events reported in >1 patient.

Table 3. Serious Treatment-Related Adverse Events Reported for Patients Treated with Paclitaxel ± Pelareorep

	Pelareorep IV Dose Group with Paclitaxel 80 mg/m² ($TCID_{50}$)	
Preferred Term	Pelareorep (N= 43) n (%)	No Pelareorep (N= 38) n (%)
Fever	3 (7.0)	0
Nausea	1 (2.3)	1 (2.6)
Vomiting	1 (2.3)	1 (2.6)
Bladder infection	1 (2.3)	0
Dyspnea	1 (2.3)	0
Febrile neutropenia	1 (2.3)	0
Flu like symptoms	1 (2.3)	0
Hypotension	1 (2.3)	0
Lung infection	1 (2.3)	0
Other infections and infestations	1 (2.3)	0
Pneumonitis	1 (2.3)	0
Dehydration	0	1 (2.6)
Diarrhea	0	1 (2.6)
Sepsis	0	1 (2.6)
Skin infection	0	1 (2.6)

Note: NCI GOG-0186H is not included in this table since a complete serious adverse event data set for this study were not provided to Oncolytics.

Studies with Carboplatin + Paclitaxel ± Pelareorep

Select Grade 1 and 2 AEs were more frequent in **R**-containing arms than in control arms in seven studies (**CPR** vs. **CP**, range in %), including: fever (86-16 vs. 22-8), chills (37-18 vs. 5-0), fatigue (86-21 vs. 51-28) and the GI-related AEs of nausea (64-30 vs. 43-30), diarrhea (54-24 vs. 24-19) and vomiting (64-16 vs. 32-14). Dyspnea was similar in both groups (21-16 vs. 21-19).

In the aggregated analysis of seven studies of **CPR**, Grade 3 or 4 TEAEs were similar in patients receiving **CP** with or without **R** (Table 4). The only difference was a higher incidence of neutropenia and leukopenia in patients receiving **R**.

Table 4. Pooled Grade 3 and 4 Treatment Emergent Adverse Events Reported for ≥ 3% of Pelareorep-Treated Patients (7 Studies): Carboplatin/Paclitaxel ± Pelareorep

Preferred Term	Pelareorep Carboplatin Paclitaxel (N= 264) n (%)	Carboplatin-Paclitaxel (N=118) n (%)
Anemia	51 (19.3)	26 (22.0)
Neutropenia	45 (17.0)	11 (9.3)
Fatigue	28 (10.6)	4 (3.4)
Hyponatremia	26 (9.8)	6 (5.1)
Neutrophil count decreased	26 (9.8)	16 (13.6)
Thrombocytopenia	20 (7.6)	8 (6.8)
Febrile neutropenia	17 (6.4)	10 (8.5)
White blood cell decreased	15 (5.7)	12 (10.2)
Hypokalemia	14 (5.3)	8 (6.8)
Platelet count decreased	14 (5.3)	10 (8.5)
Diarrhea	13 (4.9)	3 (2.5)
Dyspnea	13 (4.9)	3 (2.5)
Hypophosphatemia	12 (4.5)	2 (1.7)
Nausea	12 (4.5)	5 (4.2)
Abdominal pain	11 (4.2)	4 (3.4)
Asthenia	11 (4.2)	8 (6.8)
Hypotension	10 (3.8)	1 (0.8)
Leukopenia	10 (3.8)	0
Vomiting	10 (3.8)	3 (2.5)
Hemoglobin decreased	9 (3.4)	4 (3.4)
Hyperglycemia	9 (3.4)	9 (7.6)
Hypertension	9 (3.4)	3 (2.5)
Sepsis	8 (3.0)	1 (0.8)

The incidences of Serious TEAEs considered related to treatment by the investigator differed with respect to pyrexia, diarrhea, hypotension, dehydration, sepsis (higher in the **R** treated group) and nausea and pneumonia (higher in the **CP** treated group). The incidences of febrile neutropenia and anemia were similar in both groups (Table 5).

Table 5. Pooled Treatment-Related Serious Adverse Events Reports for ≥ 1.5% of All Treated Patients (6 Studies): Carboplatin/ Paclitaxel ± Pelareorep

Preferred Term	Pelareorep Carboplatin Paclitaxel (N= 212) n (%)	Carboplatin Paclitaxel (N=81) n (%)
Pyrexia	10 (4.7)	0
Febrile neutropenia	9 (4.2)	4 (4.9)
Anemia	7 (3.3)	2 (2.5)
Diarrhea	5 (2.4)	0
Hypotension	5 (2.4)	0
Dehydration	4 (1.9)	0
Sepsis	4 (1.9)	0
Nausea	3 (1.4)	2 (2.5)
Pneumonia	3 (1.4)	4 (4.9)

Note: NCI 8601 is not included in this table since serious adverse event data for this study were not provided to Oncolytics.

CONCLUSIONS

1. AEs reported more frequently in **R**-treated patients were Grade 1 and Grade 2 fever, chills, fatigue and the GI-related AEs of nausea, vomiting, diarrhea (and associated dehydration).
2. The frequency of Grade 3 or Grade 4 AEs induced by chemotherapy regimens do not seem to be modified by the addition of **R**.
3. Serious TEAEs like fever, GI AEs and dehydration were more common in the **R** arms. However, the incidence of SAEs due to febrile neutropenia and/or infection was similar in each group.
4. Hepatic and renal laboratory values showed no significant differences between the two groups either (data not shown). Transient Grade 3/4 laboratory abnormalities, like neutropenia and lymphopenia, may have a higher incidence when **R** is added to certain chemotherapy backbones.

REFERENCES

1. Norman, K.L. et al., 2002. Hum Gene Ther, 13(5): 641-52.
2. Gujar, S.A. and Lee, P.W., 2014. Front Oncol, 4: 77.
3. Gollamudi, R. et al., 2010. Invest New Drugs, 28(5): 641-9.
4. Bernstein, V. et al., 2017. AACR 2017 Poster, CT131.
5. Karapanagiotou, E.M. et al., 2012. Clin Cancer Res, 18(7): 2080-9.
6. Karnad, A.B. et al., 2011. AACR Meeting Abstracts 2011, 10(11 Suppl): C22.
7. Villalona-Calero, M.A. et al., 2016. Cancer, 122(6): 875-83.
8. Mita, A.C. et.al. Mol Cancer Ther 2013, 12(11 Suppl): Abstract C70.
9. Noonan, A.M. et al., 2016. Mol Ther. 24:1150–1158.

ACKNOWLEDGMENTS

ClinDatrix Statistician Julie Morduant for thorough integration of safety coding across study-specific data sets, and conduct of the statistical analysis.
Cytel Inc. Statisticians Sam Hsiao and Lingyun Liu for post-hoc analysis of the IND.213 study data.
Claire Huntley from the Granzer Group, Germany, for medical writing support.